Anti-metastatic therapy with WX-UK1 and WX-671 for the treatment of breast cancer: Phase I results and Phase II design

Carola Mala[2], Bernd Muehlenweg[2], Roman Bartz[2], Ramona Swaby[1], Paul Bevan[2], Roger B. Cohen[1], Lori J. Goldstein[1]
[1]Fox Chase Cancer Center, Philadelphia, USA; [2]Wilex AG, Munich, Germany

Introduction

The target: urokinase-type plasminogen activator

Distant metastases rather than the primary tumor itself remain the principle cause of death in patients with malignant solid tumors. uPA (urokinase-type plasminogen activator) and its inhibitor PAI-1 play a key role in tumor invasion and metastasis by being involved in the degradation of the tumor stroma, the extracellular matrix, and basement membrane. At this time uPA and PAI-1 are the only biomarkers that have been validated by the European Organization for the Treatment and Research of Cancer at the highest level of evidence (LOE I) with regard to their clinical utility in breast cancer prognosis. In the recently published "ASCO 2007 Update of Recommendations for the Use of Tumor Markers in Breast Cancer", uPA and PAI-1 were added to the list of recommended tumor markers, the system being considered a promising target for therapeutic studies.

The inhibitors: WX-UK1 and MESUPRON® (WX-671)

WX-UK1 is an inhibitor of uPA in the sub-micromolar range. WX-UK1 is not orally available and needs to be administered intravenously. WX-UK1 has been investigated in 4 phase I clinical studies, one of which is presented here.
MESUPRON® is an oral pro-drug of WX-UK1. It possesses good oral bioavailability and a molecular mass of 630 g/mol. MESUPRON® has also been investigated in four phase I clinical studies and is currently undergoing phase II development in two studies. One study evaluates MESUPRON® in pancreatic cancer, the other study (that is presented here) is in breast cancer patients.

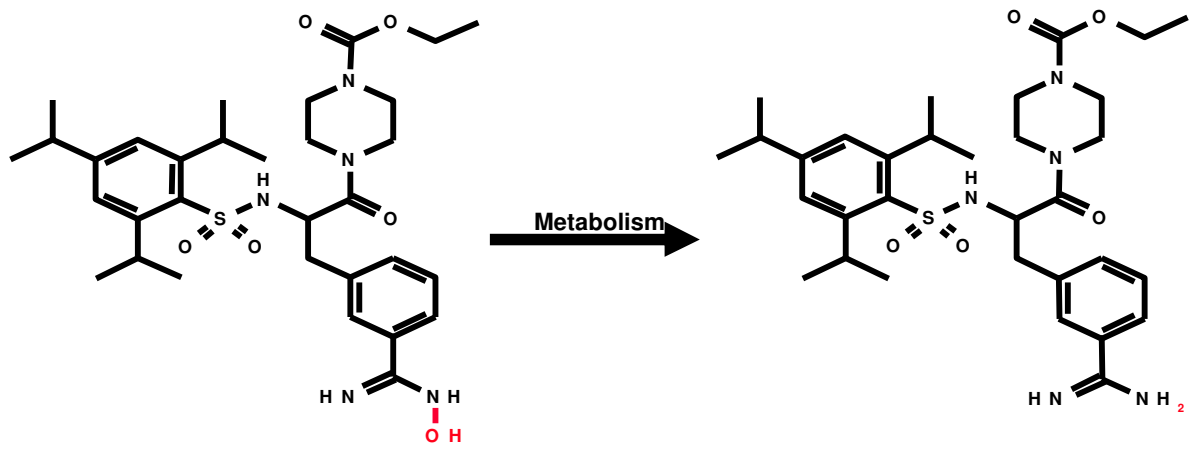
Metabolism

MESUPRON® (WX-671) **WX-UK1**

Pro-Drug Concept of **MESUPRON®**

Pharmacokinetics of WX-UK1 and WX-671

The figure below shows equivalent exposures to WX-UK1 during a treatment week with either 1.0 mg/kg WX-UK1 i.v. infused once weekly or oral MESUPRON® 200 mg once daily. The resulting plasma area under curves of WX-UK1 are similar. Daily WX-671 p.o. maintains a steady-state WX-UK1 profile sufficient to inhibit the target uPA (data not shown).

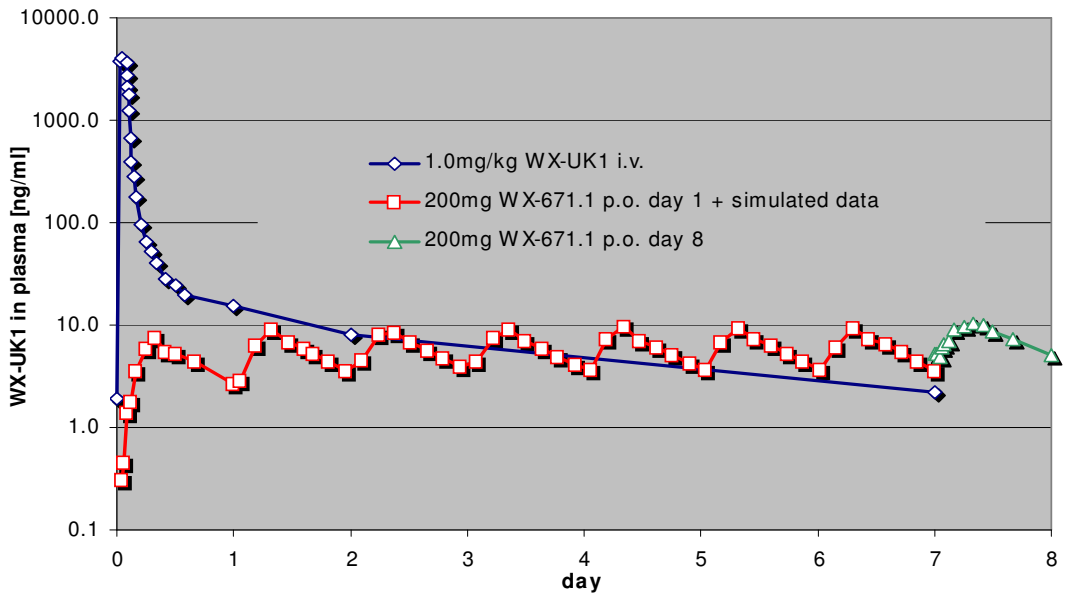
WX-UK1 in plasma [ng/ml]
10000.0
1000.0
100.0
10.0
1.0
0.1
0 1 2 3 4 5 6 7 8
day
1.0mg/kg WX-UK1 i.v.
200mg WX-671.1 p.o. day 1 + simulated data
200mg WX-671.1 p.o. day 8

Phase I design

The study aimed to evaluate WX-UK1 administered in combination with Capecitabine (Xeloda®, Roche) in patients with advanced solid tumors. Escalating doses of WX-UK1 were given weekly (day 2, day 9, day 16) in combination with Capecitabine (day 1-14) in 3-week treatment cycles until disease progression or unacceptable toxicity occurs.
The pharmacokinetic properties of each drug were measured as single agents and in combination, with day 1 being the Capecitabine alone day, day 9 the combination day, and day 16 the WX-UK1 alone study day.
Capecitabine was given orally at a fixed dose of 1000 mg/m^2 every 12 hours, whereas WX-UK1 was dose-escalated starting at 0.3 mg/kg, and administered as a 2-hour intravenous constant rate infusion. The study was designed to investigate safety, tolerability, pharmacokinetics and pharmacodynamics of the drug combination.

Phase I statistics

- 38 patients were screened – 30 patients were allocated to treatment
- All patients with metastatic, incurable tumors and no further standard treatment options
- 23 patients completed at least one cycle
- 6 different dose levels of WX-UK1 between 0.3 and 2.8 mg/kg b.w.
- Maximum tolerated dose not reached, no WX-UK1 related dose limiting toxicities
- Tumor origin (treated patients):
 - Breast (10)
 - Pancreas (4)
 - Other (14)
- On average 4.8 treatment cycles per patient
- Four patients with $\geq$ 11 courses (range 11-15), 3 of these 4 with metastatic breast cancer
- Total number of 110 treatment courses were given

Phase I Safety and Pharmacokinetics

- WX-UK1 in combination with Capecitabine was well-tolerated
- The incidence of Adverse Events was unrelated to WX-UK1 dose level
- No Serious Adverse Events were related to WX-UK1
- No WX-UK1 related acute systemic, allergic, or local intolerance reaction
- Effect on coagulation at C_{max} was transient, completely reversible and of no clinical relevance
- Adverse Events mostly GI, respiratory, skin & subcutaneous, or lab events, no explicit dose or infusion relationship to WX-UK1 detected
- No apparent changes in the frequencies or intensities of Capecitabine related side effects reported
- WX-UK1 plasma Area Under Curve appear to be dose-linear
- No reciprocal drug-drug interactions WX-UK1 and Capecitabine observed

Phase I Efficacy

Out of twenty evaluable patients
- 3 patients experienced partial responses
- 9 patients experienced stable disease with an average of 3.4 months (range 1.5 to 8.3 months)
- 8 patients had progressive disease after treatment cycle 2
- 2 patients with CR of liver metastases (both MBC patients)

Overall, the number of cycles per patient was higher than expected prior to the study in this heavily pretreated patient population which may suggest clinical benefit.

Phase I Conclusions

No WX-UK1 related dose limiting toxicities were observed during the entire treatment period, and no maximum tolerated dose could be identified. This demonstrates that the combination was well tolerated by all patients for the duration of the study.
WX-UK1 has dose linear pharmacokinetic properties over the entire dose range and there is no significant drug-drug interaction between Capecitabine and WX-UK1.
Some patients showed a partial response or stable disease over a longer period of time during the study. This is encouraging as these patients all had advanced, metastatic tumors, and no standard efficacious treatment existed for them.
Data from three further Phase I studies with WX-671 support a switch from i.v. WX-UK1 to oral WX-671 for the Phase II study in metastatic breast cancer patients.

Phase II design **ClinicalTrials.gov Identifier: NCT00615940**

Design

- Phase II, double-blind, randomized
- Daily 200 mg of oral WX-671 plus Capecitabine vs. Capecitabine;
- 50 evaluable patients per arm
- First-line HER2-negative metastatic breast cancer

Endpoints

- Progression-free survival
- Objective response rate
- Overall survival
- Safety
- Pharmacokinetics

Support for this study is provided by the U.S. Army Medical Research and Material Command under DAMD17-03-1-0634

www.wilex.com